Facilitated Immunoglobulin Administration Registry and Outcomes Study (FIGARO): Interim Results in Patients With Secondary Immunodeficiency Diseases (SID)

P071

Maria Dimou,[1] Dörte Huscher,[2] Karen Cheng,[3] Corinna Hermann,[3] David Pittrow,[4] Matthaios Speletas[5]

[1]First Department of Propaedeutic Internal Medicine, National & Kapodistrian University of Athens Medical School, General Hospital "LAIKO," Athens, Greece;
[2]Institute of Biometry and Clinical Epidemiology, Charité Universitätsmedizin Berlin, Berlin, Germany; [3]Baxalta Innovations GmbH, a Takeda company, Vienna, Austria;
[4]Institute for Clinical Pharmacology, Medical Faculty, Technical University of Dresden, Dresden, Germany; GWT-TUD GmbH, Pharmacoepidemiology, Dresden, Germany;
[5]School of Health Sciences Faculty of Medicine, Department of Immunology and Histocompatibility, Medical School Campus - University of Thessaly, Thessaly, Greece

Introduction

Background

- Secondary immunodeficiency diseases (SID) are a frequent complication of hematologic malignancies such as chronic lymphocytic leukemia (CLL) or multiple myeloma (MM) and associated treatments[1]
 - Patients who have SID are at an increased risk of infections, which are a major cause of morbidity and mortality[2]
- Several national guidelines recommend the use of immunoglobulin replacement therapy (IGRT) to reduce the incidence of infections among patients who have SID and repeated infections despite using prophylactic antibiotic therapy[1]
 - IGRT can be administered intravenously (IVIG) or subcutaneously (SCIG). Unlike IVIG, conventional SCIG (cSCIG) allows for self-administration at home with better systemic tolerability
 - However, SCIG is limited by the volume that can be infused, necessitating more frequent infusions and multiple infusion sites[3,4]

Facilitated Subcutaneous Immunoglobulin

- Facilitated SCIG (fSCIG; Baxalta US Inc., a Takeda company) is a dual-vial unit of immunoglobulin G (IgG) 10% and recombinant human hyaluronidase (rHuPH20)[5]
 - rHuPH20 depolymerizes hyaluronan in the extracellular matrix, resulting in a transient and local increase in subcutaneous tissue permeability, allowing for larger volumes / infusion site relative to cSCIG[4] (**Figure 1**)

Figure 1. rHuPH20 Mechanism of Action Allows Increased Volume and Flow Rate of IG Infusion Into the Subcutaneous Tissue

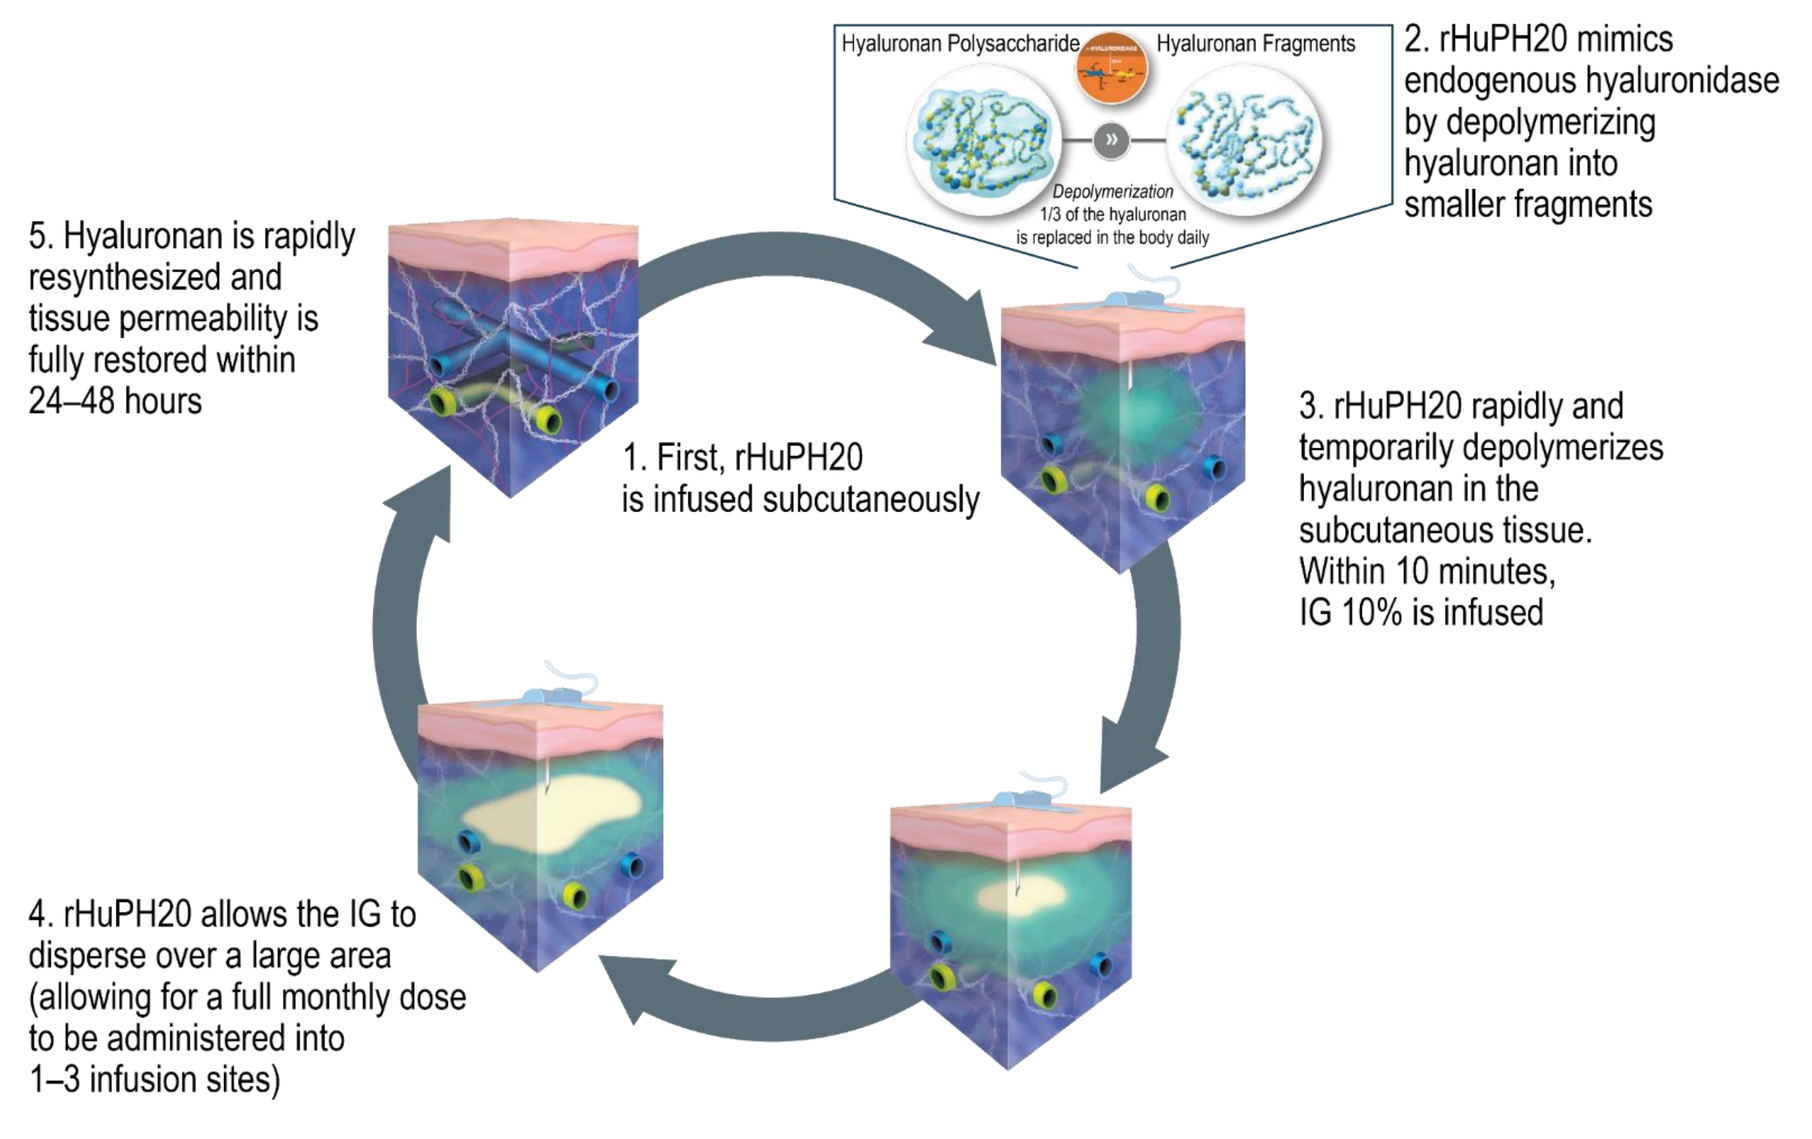

Hyaluronan Polysaccharide
Hyaluronan Fragments
Depolymerization 1/3 of the hyaluronan is replaced in the body daily
2. rHuPH20 mimics endogenous hyaluronidase by depolymerizing hyaluronan into smaller fragments
5. Hyaluronan is rapidly resynthesized and tissue permeability is fully restored within 24–48 hours
1. First, rHuPH20 is infused subcutaneously
3. rHuPH20 rapidly and temporarily depolymerizes hyaluronan in the subcutaneous tissue. Within 10 minutes, IG 10% is infused
4. rHuPH20 allows the IG to disperse over a large area (allowing for a full monthly dose to be administered into 1–3 infusion sites)

IG, immunoglobulin; rHuPH20, recombinant human hyaluronidase.

- fSCIG is approved in Europe for the treatment of primary immunodeficiency diseases (PID) and SID[a5]
 - In a pivotal phase 3 study of patients with PID (NCT00814320), fSCIG was effective, safe, and bioequivalent to IVIG at the same administration intervals, with fewer systemic reactions. Annual overall infection rates during treatment with IVIG and fSCIG were 4.51 and 2.97 events / patient-years, respectively[6]
- Additional data on fSCIG use in patients with SID are needed

Objective

- The objective of this analysis is to assess the use of fSCIG in routine clinical practice in patients with SID

Methods

- FIGARO (NCT03054181) is a multicenter, prospective observational study conducted in Europe under the auspices of the European Society for Immunodeficiencies
- Patients are eligible for inclusion if they:
 - Received ≥ 1 fSCIG infusion for PID or SID
 - Provide informed consent
- An interim analysis (data cut-off 12 March 2020) was conducted on data collected at inclusion visits in the subset of patients with SID
 - Parameters assessed included:
 - Patient characteristics and medical history
 - fSCIG utilization patterns at inclusion visit
 - IgG trough levels at inclusion visit
 - fSCIG tolerability (local or systemic adverse drug reactions [ADRs])
- Patients will be followed for up to 3 years

Results

Patients and Baseline Characteristics

- Of the 154 patients enrolled at the data cutoff date, 31 received fSCIG for SID and were included in the analysis
 - Nineteen (61.3%) patients were male, and 29 (93.5%) were Caucasian / White
 - The mean (standard deviation; SD) age was 61.4 (17.8) years (range: 3–88 years)
 - The most common SID diagnoses were CLL (64.5%) and non-Hodgkin lymphoma (19.4%) (**Figure 2**)

Figure 2. SID Diagnoses at Baseline (Number of Patients)

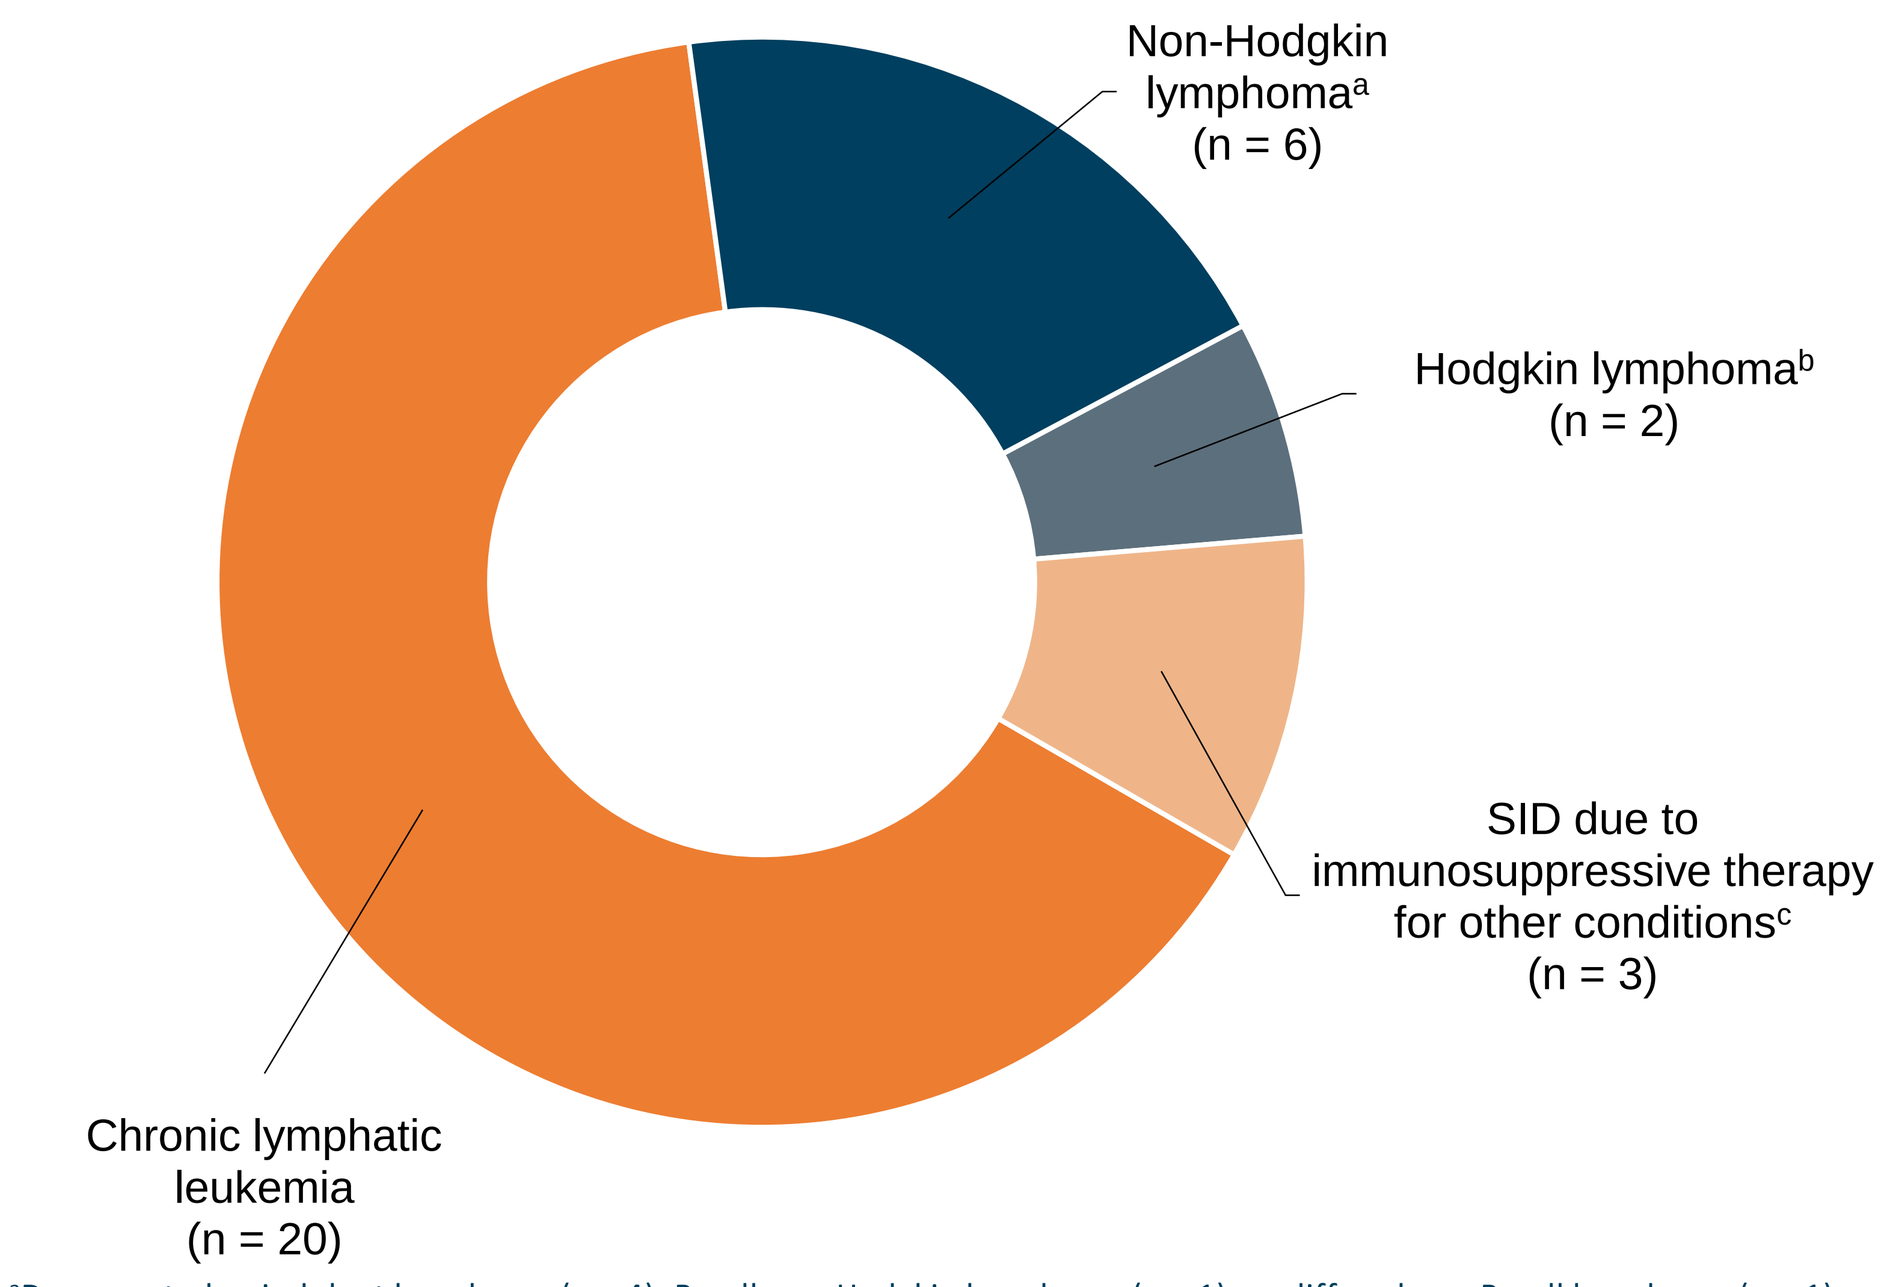

Non-Hodgkin lymphoma[a] (n = 6)
Hodgkin lymphoma[b] (n = 2)
SID due to immunosuppressive therapy for other conditions[c] (n = 3)
Chronic lymphatic leukemia (n = 20)

[a]Documented as indolent lymphoma (n = 4), B-cell non-Hodgkin lymphoma (n = 1), or diffuse large B-cell lymphoma (n = 1).
[b]Documented as Hodgkin lymphoma (n = 1) or Hodgkin lymphoma after autologous transplantation (n = 1).
[c]Documented as lung transplantation with rituximab therapy (n = 1), rheumatoid arthritis with sulfasalazine therapy (n = 1), and liver disease with corticosteroids and tacrolimus therapy (n = 1).

SID, secondary immunodeficiency diseases.

- At baseline, 28 (90.3%) patients were receiving chemotherapy and/or immunosuppressive or supportive therapy (**Figure 3**)
 - The most common chemotherapy / immunosuppressive therapies were venetoclax and ibrutinib
 - The most common supportive therapies were *Pneumocystis jiroveci* pneumonia prophylaxis, virostatics, and antibiotics

Figure 3. Concomitant Chemotherapies, Immunosuppressive Therapies, and Supportive Therapies at Baseline (Number of Patients)

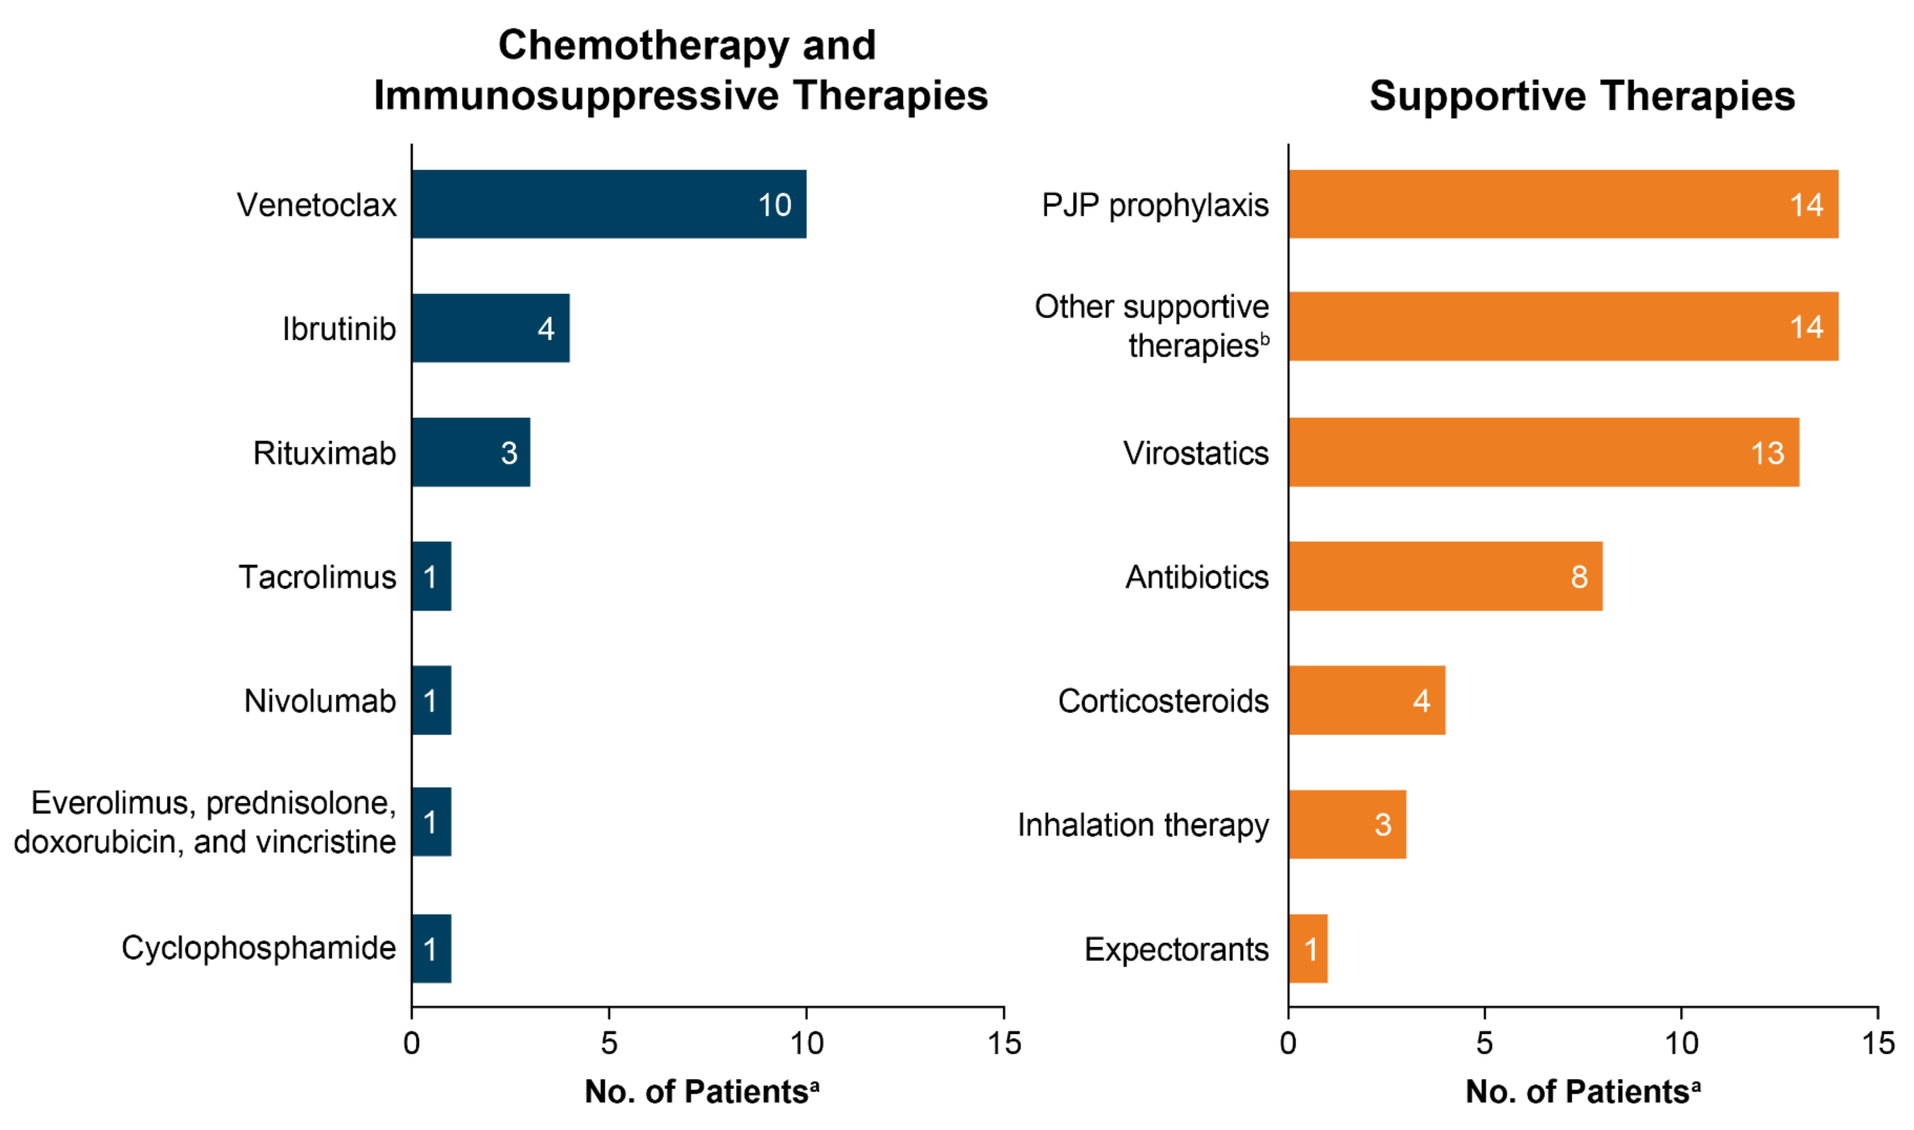

Chemotherapy and Immunosuppressive Therapies
Venetoclax 10
Ibrutinib 4
Rituximab 3
Tacrolimus 1
Nivolumab 1
Everolimus, prednisolone, doxorubicin, and vincristine 1
Cyclophosphamide 1
0 5 10 15
No. of Patients[a]
Supportive Therapies
PJP prophylaxis 14
Other supportive therapies[b] 14
Virostatics 13
Antibiotics 8
Corticosteroids 4
Inhalation therapy 3
Expectorants 1
0 5 10 15
No. of Patients[a]

[a]Patients could have received > 1 therapy.
[b]Other supportive therapies reported in > 1 patient were allopurinol (n = 7) and aciclovir (n = 5).
PJP, *Pneumocystis jiroveci* pneumonia.

Infusion Parameters / fSCIG Administration

- Infusion parameters at inclusion visit are described in **Table 1** and **Figure 4**
 - Mean (SD) monthly fSCIG dose was 26.6 (6.1) g
 - Most patients self-administered fSCIG (58.1%) and infused at home (61.3%)
 - Patients predominantly (67.7%) received fSCIG treatment every 4 weeks
 - All but 1 patient (96.3%) used a single infusion site
 - Only 1 of 31 patients required premedication
 - No technical problems were reported, and all patients received the full fSCIG dose as planned on the scheduled date (93.1%) or within ±1–3 days of the scheduled date (6.9%)

Table 1. fSCIG Infusion Parameters (Inclusion Visit)

Parameter[a]	
Monthly fSCIG dose, mean (SD), g	26.6 (6.1)
Infusion volume, median (range), mL	300.0 (100.0–300.0)
Maximum infusion rate, median (range), mL/hr	290.0 (180.0–300.0)

[a]Of patients with available data.
fSCIG, facilitated subcutaneous immunoglobulin; SD, standard deviation.

- The mean (SD) IgG serum trough level at the inclusion visit was 5.9 (1.9) g/L

Tolerability

- One patient reported a local ADR (erythema), and 1 patient had a systemic ADR (severe headache) at the inclusion visit

Figure 4. fSCIG Administration (Number of Patients; Inclusion Visit)

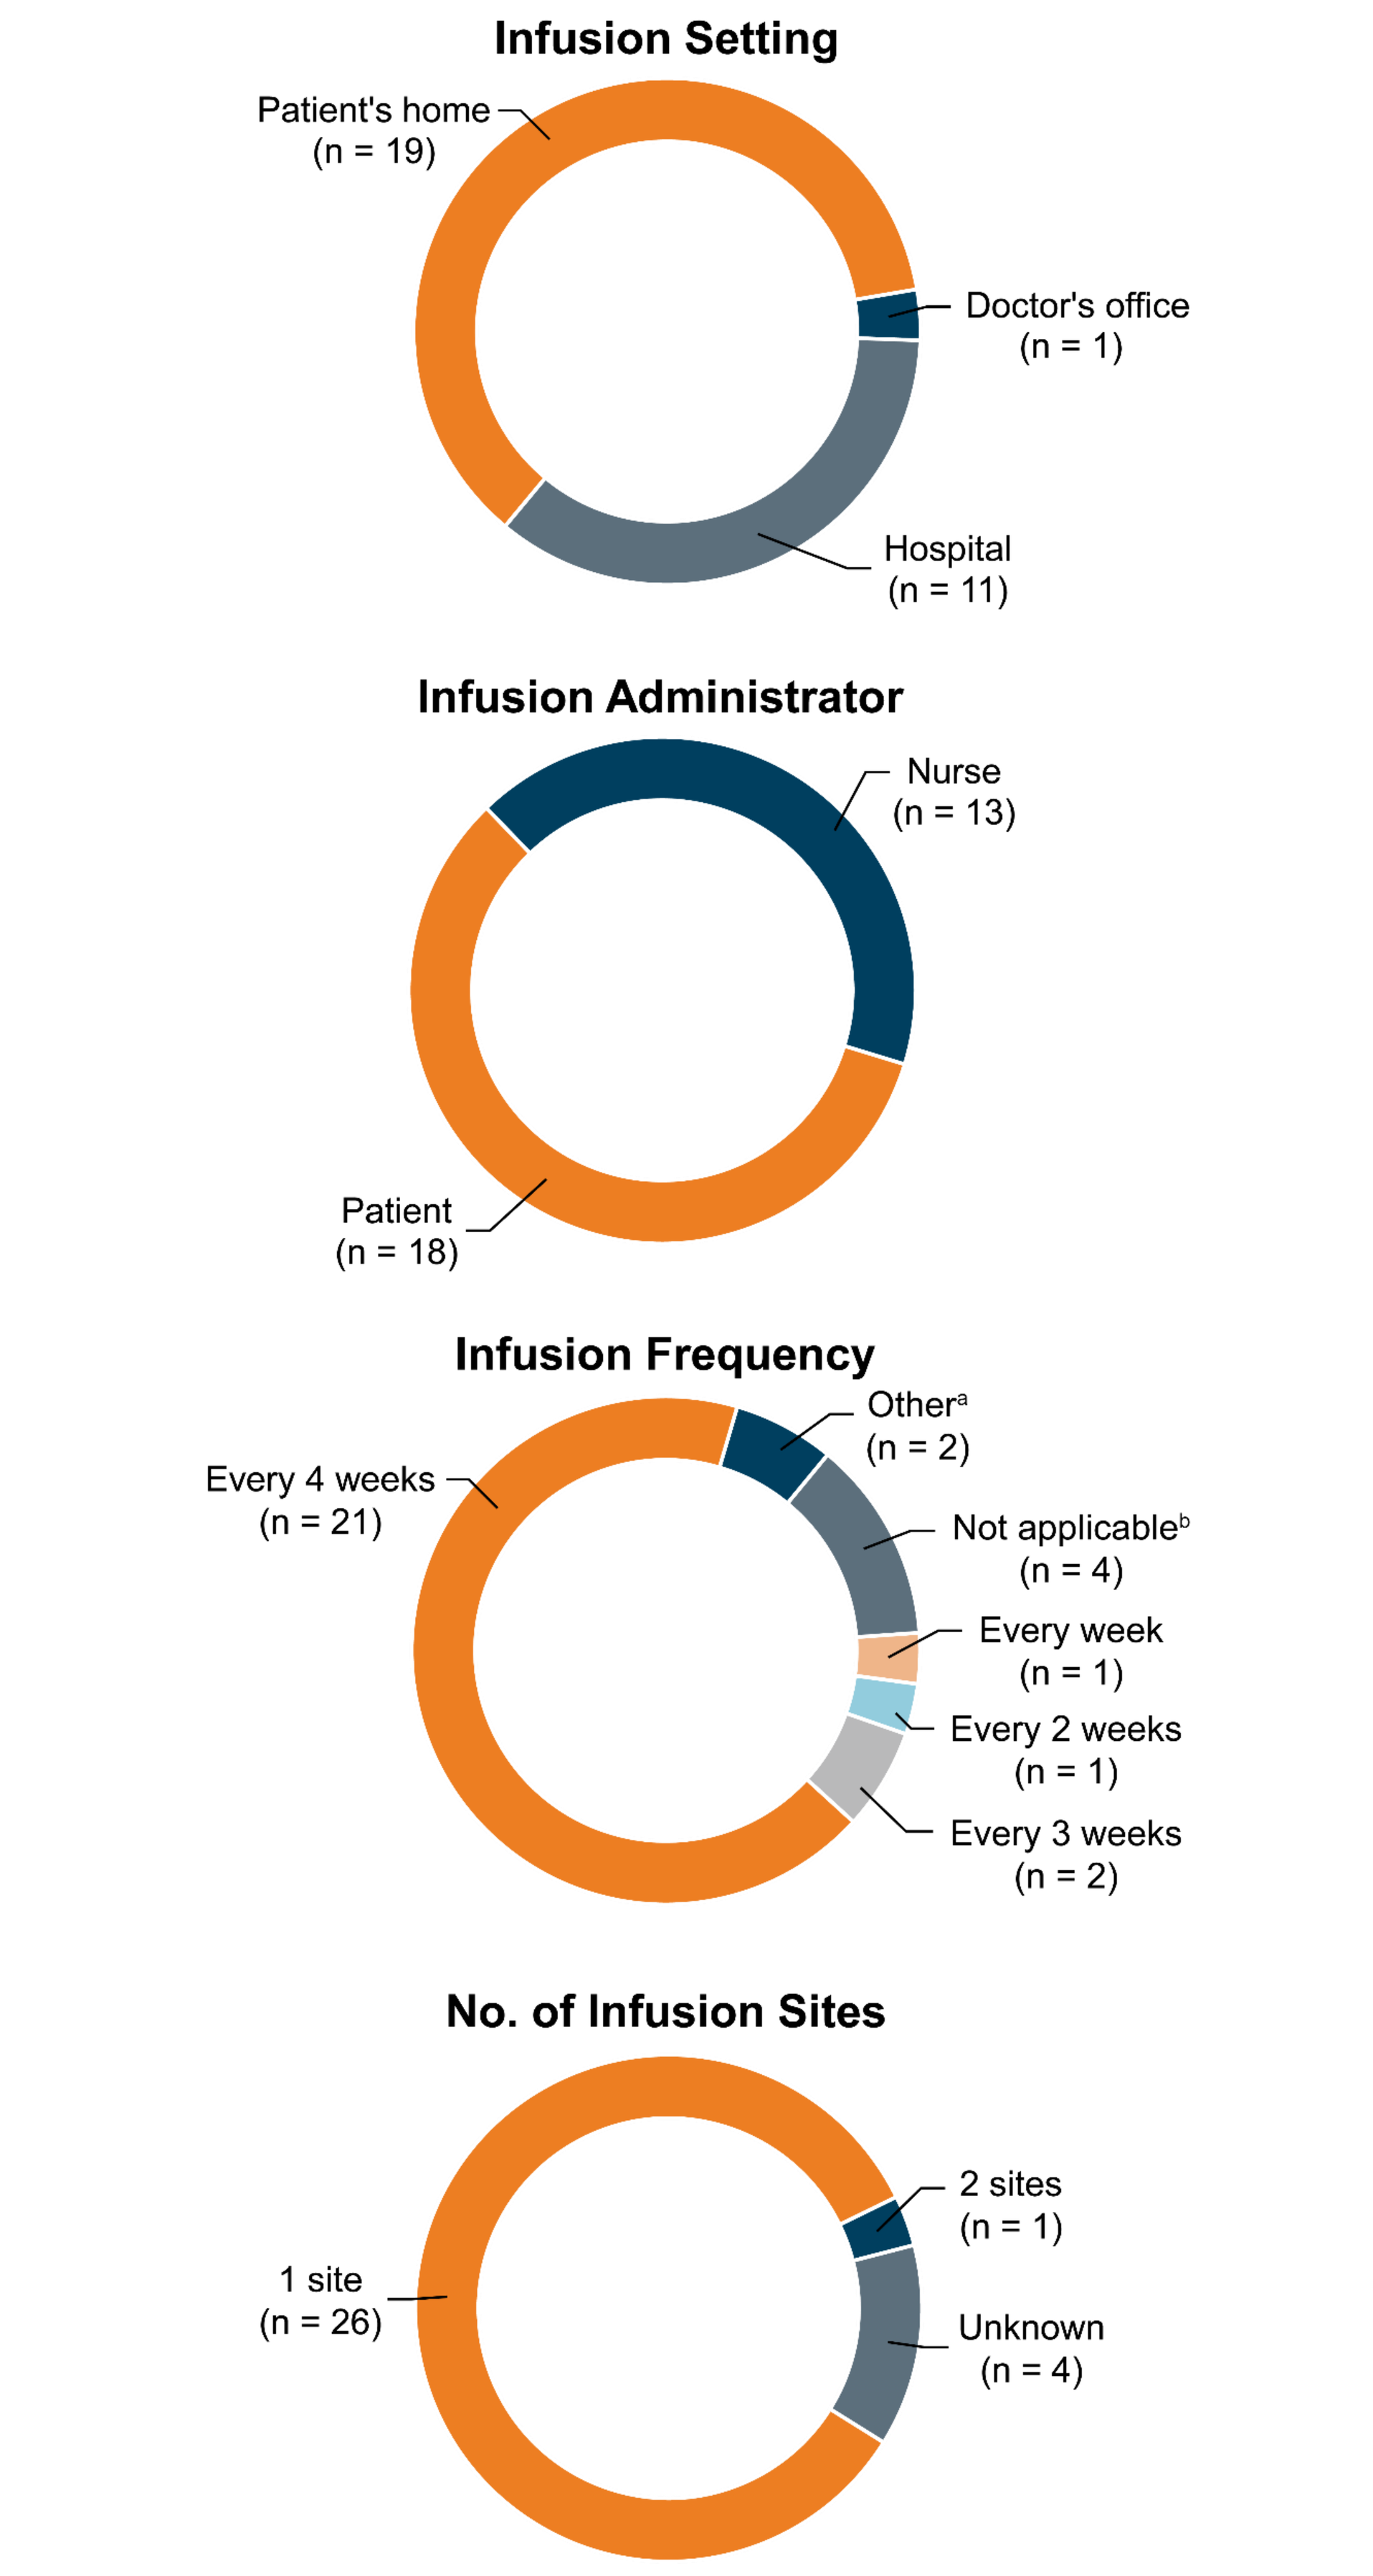

Infusion Setting
Patient's home (n = 19)
Doctor's office (n = 1)
Hospital (n = 11)
Infusion Administrator
Nurse (n = 13)
Patient (n = 18)
Infusion Frequency
Other[a] (n = 2)
Every 4 weeks (n = 21)
Not applicable[b] (n = 4)
Every week (n = 1)
Every 2 weeks (n = 1)
Every 3 weeks (n = 2)
No. of Infusion Sites
2 sites (n = 1)
1 site (n = 26)
Unknown (n = 4)

[a]Every 8 weeks (n = 1) or 12 weeks (n = 1).
[b]Patients had received only 1 fSCIG infusion to date.
fSCIG, facilitated subcutaneous immunoglobulin.

Conclusions

- **In patients with SID primarily due to hematologic malignancies, fSCIG provided the flexibility for subcutaneous infusion at home or in the hospital setting or doctor's office, either by the patient or a nurse**
- **The dosing schedule similarly allowed for flexibility, although most infusions were administered every 4 weeks, with > 95% of patients using 1 infusion site**
- **fSCIG tolerability for patients with SID in clinical practice was consistent with previous observations in patients with PID**
- **Patient observation is ongoing**

[a]In patients who suffer from severe or recurrent infections, ineffective antimicrobial treatment and either proven specific antibody failure[b] or serum IgG level of <4 g/L

[b]Defined as failure to mount at least a 2-fold rise in IgG antibody titer to pneumococcal polysaccharide and polypeptide antigen vaccines

References

1. Patel SY, et al. *Front Immunol*. 2019;10:33.
2. Duraisingham SS, et al. *Expert Rev Clin Immunol*. 2014;10(5):583–591.
3. Compagno N, et al. *Haematologica*. 2014;99(6):1101–1106.
4. Wasserman RL. *Immunotherapy*. 2017;9(12):1035–1050.
5. HyQvia 100 mg/mL solution for infusion for subcutaneous use. Summary of Product Characteristics. Baxter Innovations GmbH, Vienna, Austria, 2020.
6. Wasserman RL, et al. *J Allergy Clin Immunol*. 2012;130(4):951–957.

Acknowledgments

The authors thank the patients who participated in this trial, any caregivers, study-site personnel, and the investigators. Medical writing services were provided by Amanda Borrow, PhD, of Oxford PharmaGenesis Inc., and funded by Baxalta US Inc., a Takeda company. This study was funded by Baxalta Innovations GmbH, a Takeda company. GWT-TUD GmbH, Dresden, Germany, is the study sponsor.

Author disclosures

MD has nothing to disclose. **DH** has received travel compensation from Actelion Pharmaceuticals, Switzerland; Boehringer-Ingelheim, Germany; and Shire. **CH** is an employee of Baxalta Innovations GmbH, a member of the Takeda group of companies, and a Takeda stock owner. **DP** reports personal fees from Actelion, Bayer, Aspen, Boehringer Ingelheim, Sanofi, Biogen, and MSD, outside the submitted work. **DP** has acted as consultant for Baxalta. **MS**'s institution has received a research grant from Shire.

Previously presented at the European Hematology Society Annual Congress; June 11–14, 2020

Mobile and environmentally friendly e-posters
2 ways to access the poster
Access online
www.shirecongressposters.com/929722
No personal information is stored - standard charges may apply.
Scan QRcode

This poster is intended for healthcare providers

Presented at: XVI SIES National Congress – Italian Society of Experimental Hematology; October 15–17, 2020; Naples, Italy